Glycopyrrolate and Formoterol Fumarate Monotherapy and Combination Metered Dose Inhalers with High Dose Uniformity and Stable Aerosol Properties

R. Vehring, M. Hartman, R. Schultz*, V. Joshi, M. Sommerville, H. Cummings, A. Smith, M. Golden, C. Reisner, S. Dwivedi
Redwood City CA/US

Abstract

Product performance test data are presented on Glycopyrrolate (GP), a LAMA, and Formoterol Fumarate (FF), a LABA, and their combination in Pearl's HFA MDI format (GP MDI, FF MDI, and GP/FF combination MDI). GP and FF MDIs consistently deliver 18 and 2.4 µg GP and FF per actuation, respectively, with over 50% of the delivered dose in a particle size range suitable for uniform deposition in human airways. Their aerodynamic particle size distributions show excellent long term stability when stored at refrigerated (2-8°C), room temperature (25°C/60% RH) or stressed (40°C/75% RH) conditions. The delivered doses and aerosol properties of GP MDIs remain unchanged upon repeat thermal excursions between -5°C and 40°C for several weeks, demonstrating the robustness of Pearl's novel HFA MDI suspensions. GP/FF MDIs also show excellent stability without any physical or chemical interaction between the two actives under a broad range of test conditions. The overall performance attributes for the two drugs in GP/FF MDI combination remain unchanged from the monotherapy GP and FF MDIs.

Introduction

Metered dose inhalers (MDIs) are the most widely used inhaled dosage form, yet important therapies are not available in this format due to technical problems encountered in developing stable MDIs with hydrofluoroalkane (HFA) propellants across a broad range of drugs. Long acting muscarinic antagonists (LAMA), long acting β_2 adrenergic receptor-agonists (LABA), and their combinations, for example, have been particularly difficult to formulate as suspension MDIs due to their very low doses and physico-chemical instability in HFAs. Pearl's novel suspension-based MDI technology overcomes these difficulties by the use of porous particles that suspend well in HFAs, conforming to the most stringent regulatory requirements for monotherapy and combination products alike.

Separate single-dose, dose-ranging studies in patients with COPD have been completed on GP MDI (PT001; ATS poster F87) and FF MDI (PT005; ATS poster F95), and GP/FF combination MDI have been evaluated in a healthy volunteer study (PT003; ATS poster F92).

Pearl is well positioned to progress these products into chronic dosing studies, as well as develop other drugs in its superior MDI platform to offer patients the same dosage form across a broad range of therapeutics.

Materials and Methods

Pearl's porous particle based suspension MDIs utilize spray dried particles made from phospholipid and calcium chloride. These porous particles form highly uniform and stable suspensions across a wide range of actives and their combinations. Very low dose potent APIs are also successfully being developed with this technology.

Pearl's MDIs are manufactured with HFA and porous particles using standard valves, cans, and actuators.

Materials and Methods

Drugs and combinations can be spray dried with the porous particles or included as microcrystals with no further modification and without any additional excipients.

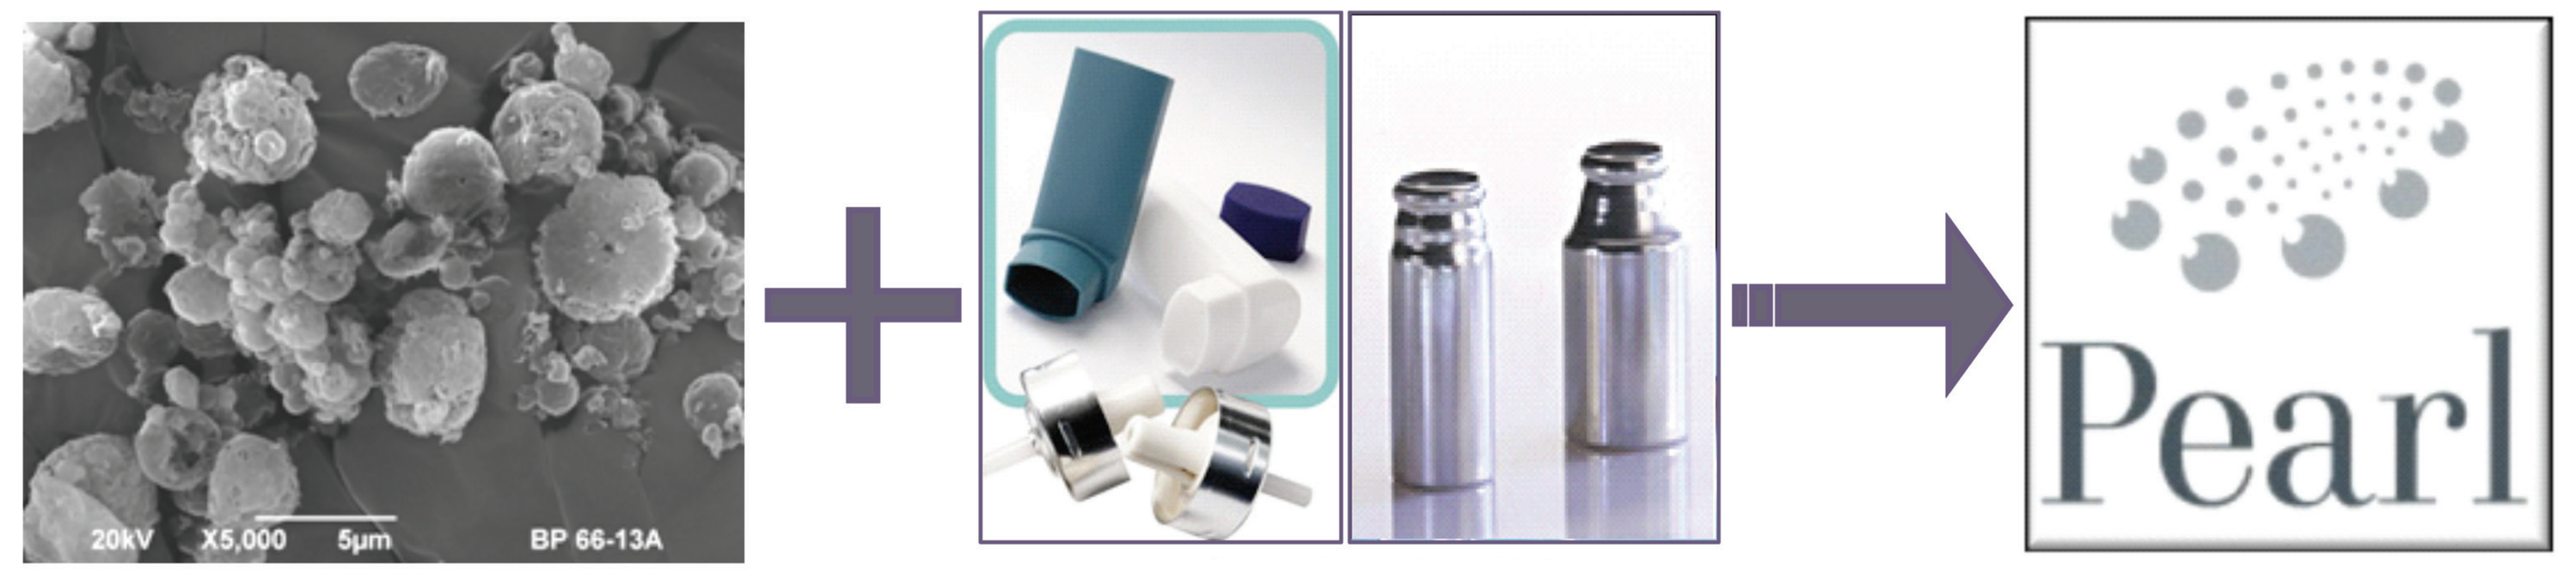
20kV X5,000 5µm BP 66-13A
Pearl

Pearl proprietary engineered particles and formulations — Standard MDI components[1] — MDI Products

Cascade impaction (using Next Generation Impactor), delivered dose uniformity (beginning, middle and end of canister life) and degradation products data were collected for inhalers placed on stability. Suspension quality was also assessed over time after shaking by visual observation of formulations filled in glass vials.

Results

Superior Suspension Stability Ensures Uniform Dosing

Conventional MDI[2]

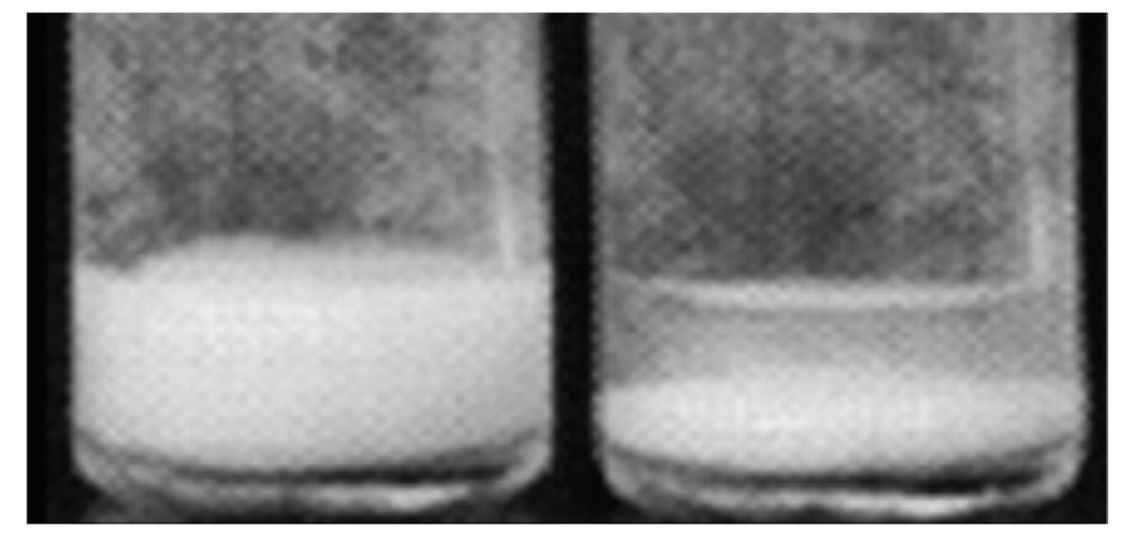

0 seconds ➔ 5 seconds

Pearl MDI

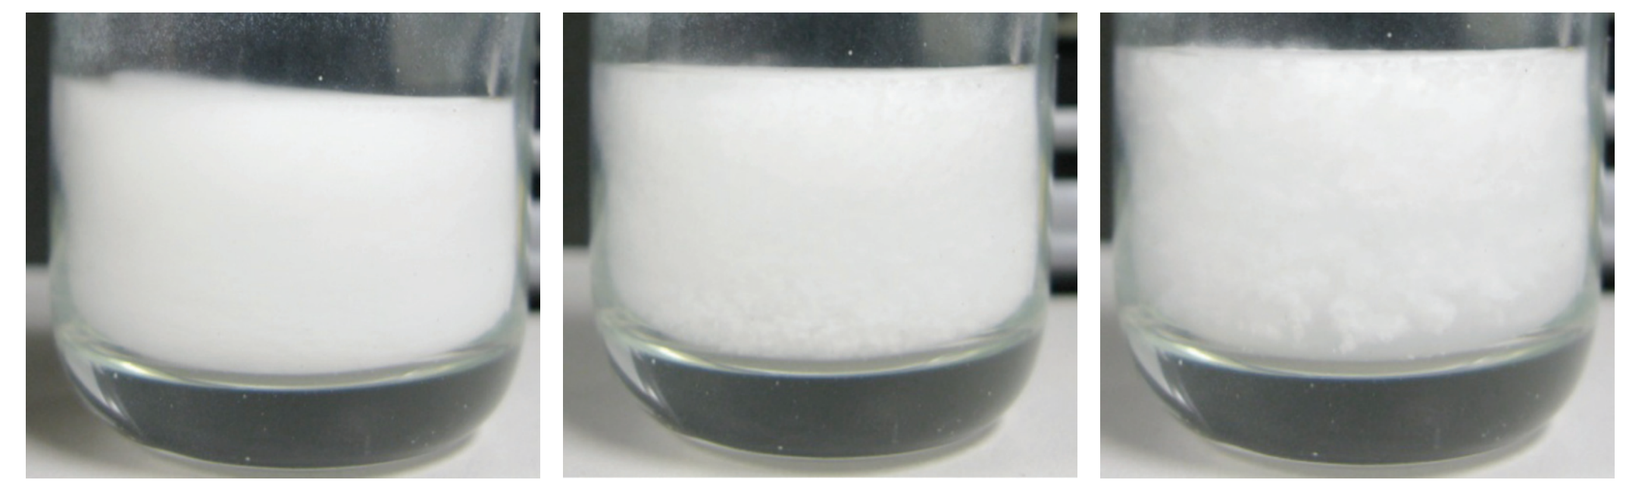

0 seconds ➔ 15 seconds ➔ 30 seconds

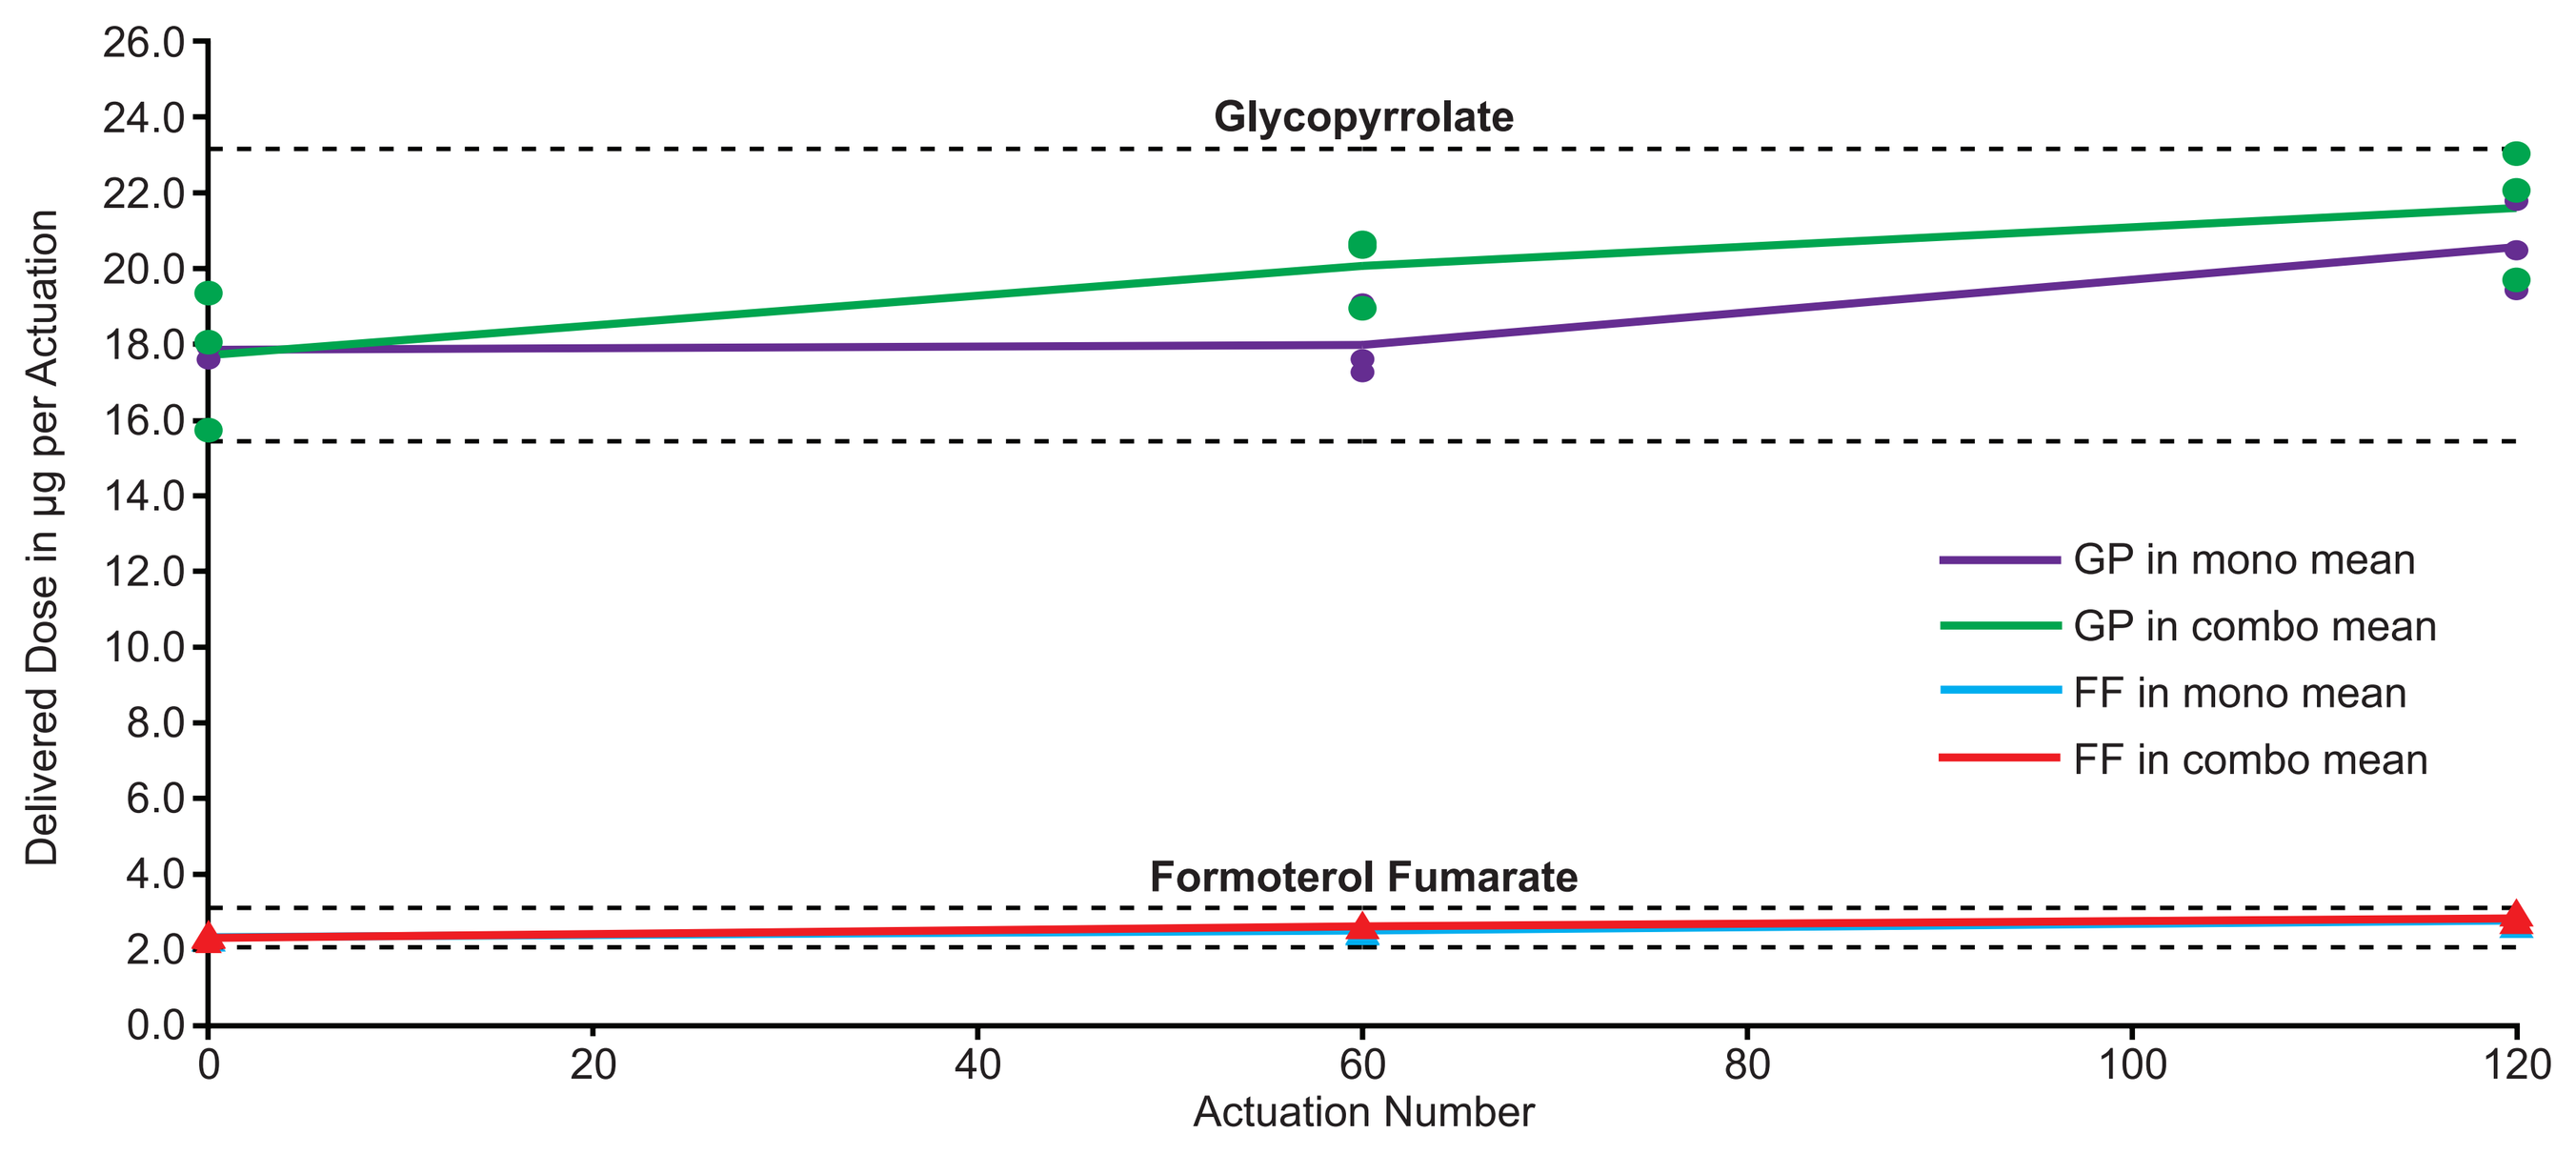
Glycopyrrolate
Formoterol Fumarate
Delivered Dose in µg per Actuation
Actuation Number
GP in mono mean
GP in combo mean
FF in mono mean
FF in combo mean

Figure 1. GP MDI (18 µg), FF MDI (2.4 µg) and GP/FF MDI (18/2.4 µg) Dosing (±20% of mean doses shown)

Results

Ability to Formulate Very Low Doses

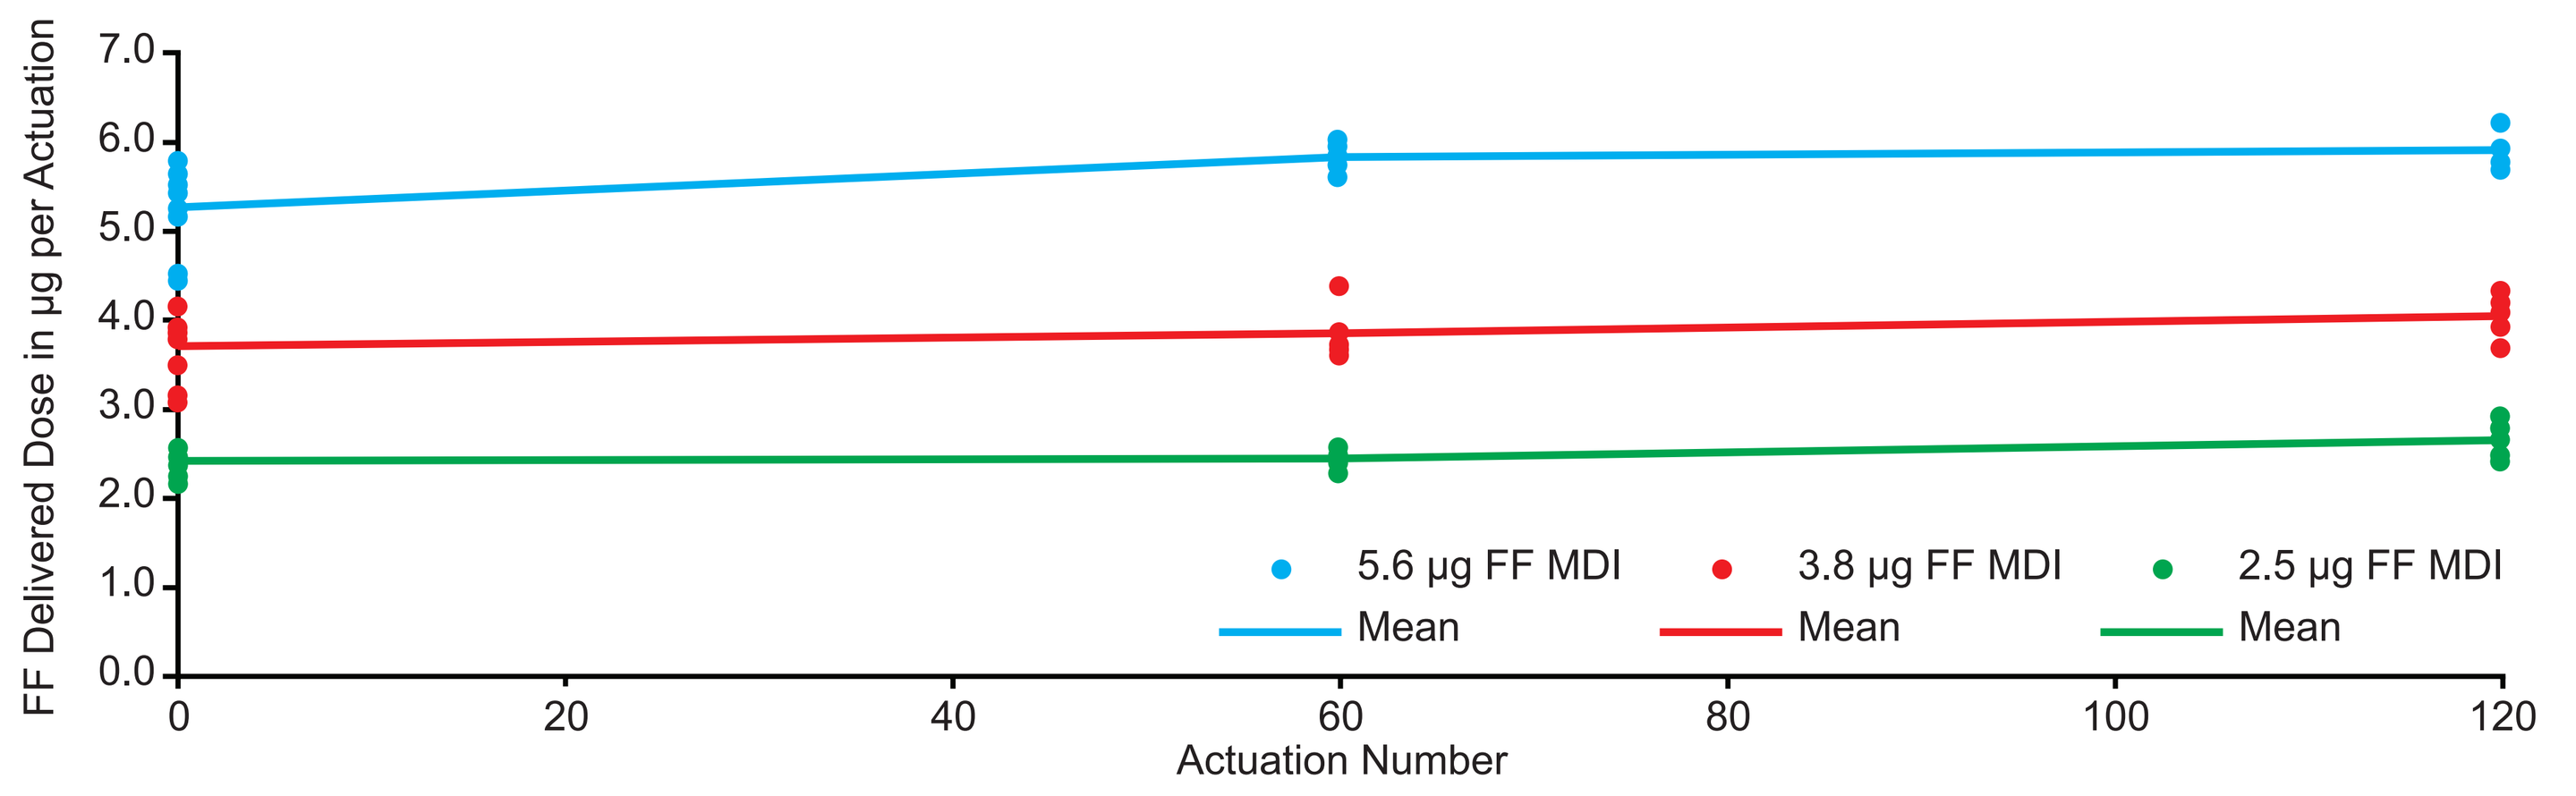
FF Delivered Dose in µg per Actuation
Actuation Number
5.6 µg FF MDI
3.8 µg FF MDI
2.5 µg FF MDI
Mean

Figure 2. Low Strength FF MDI Delivered Dose Uniformity

Highly Consistent and Robust Aerosols

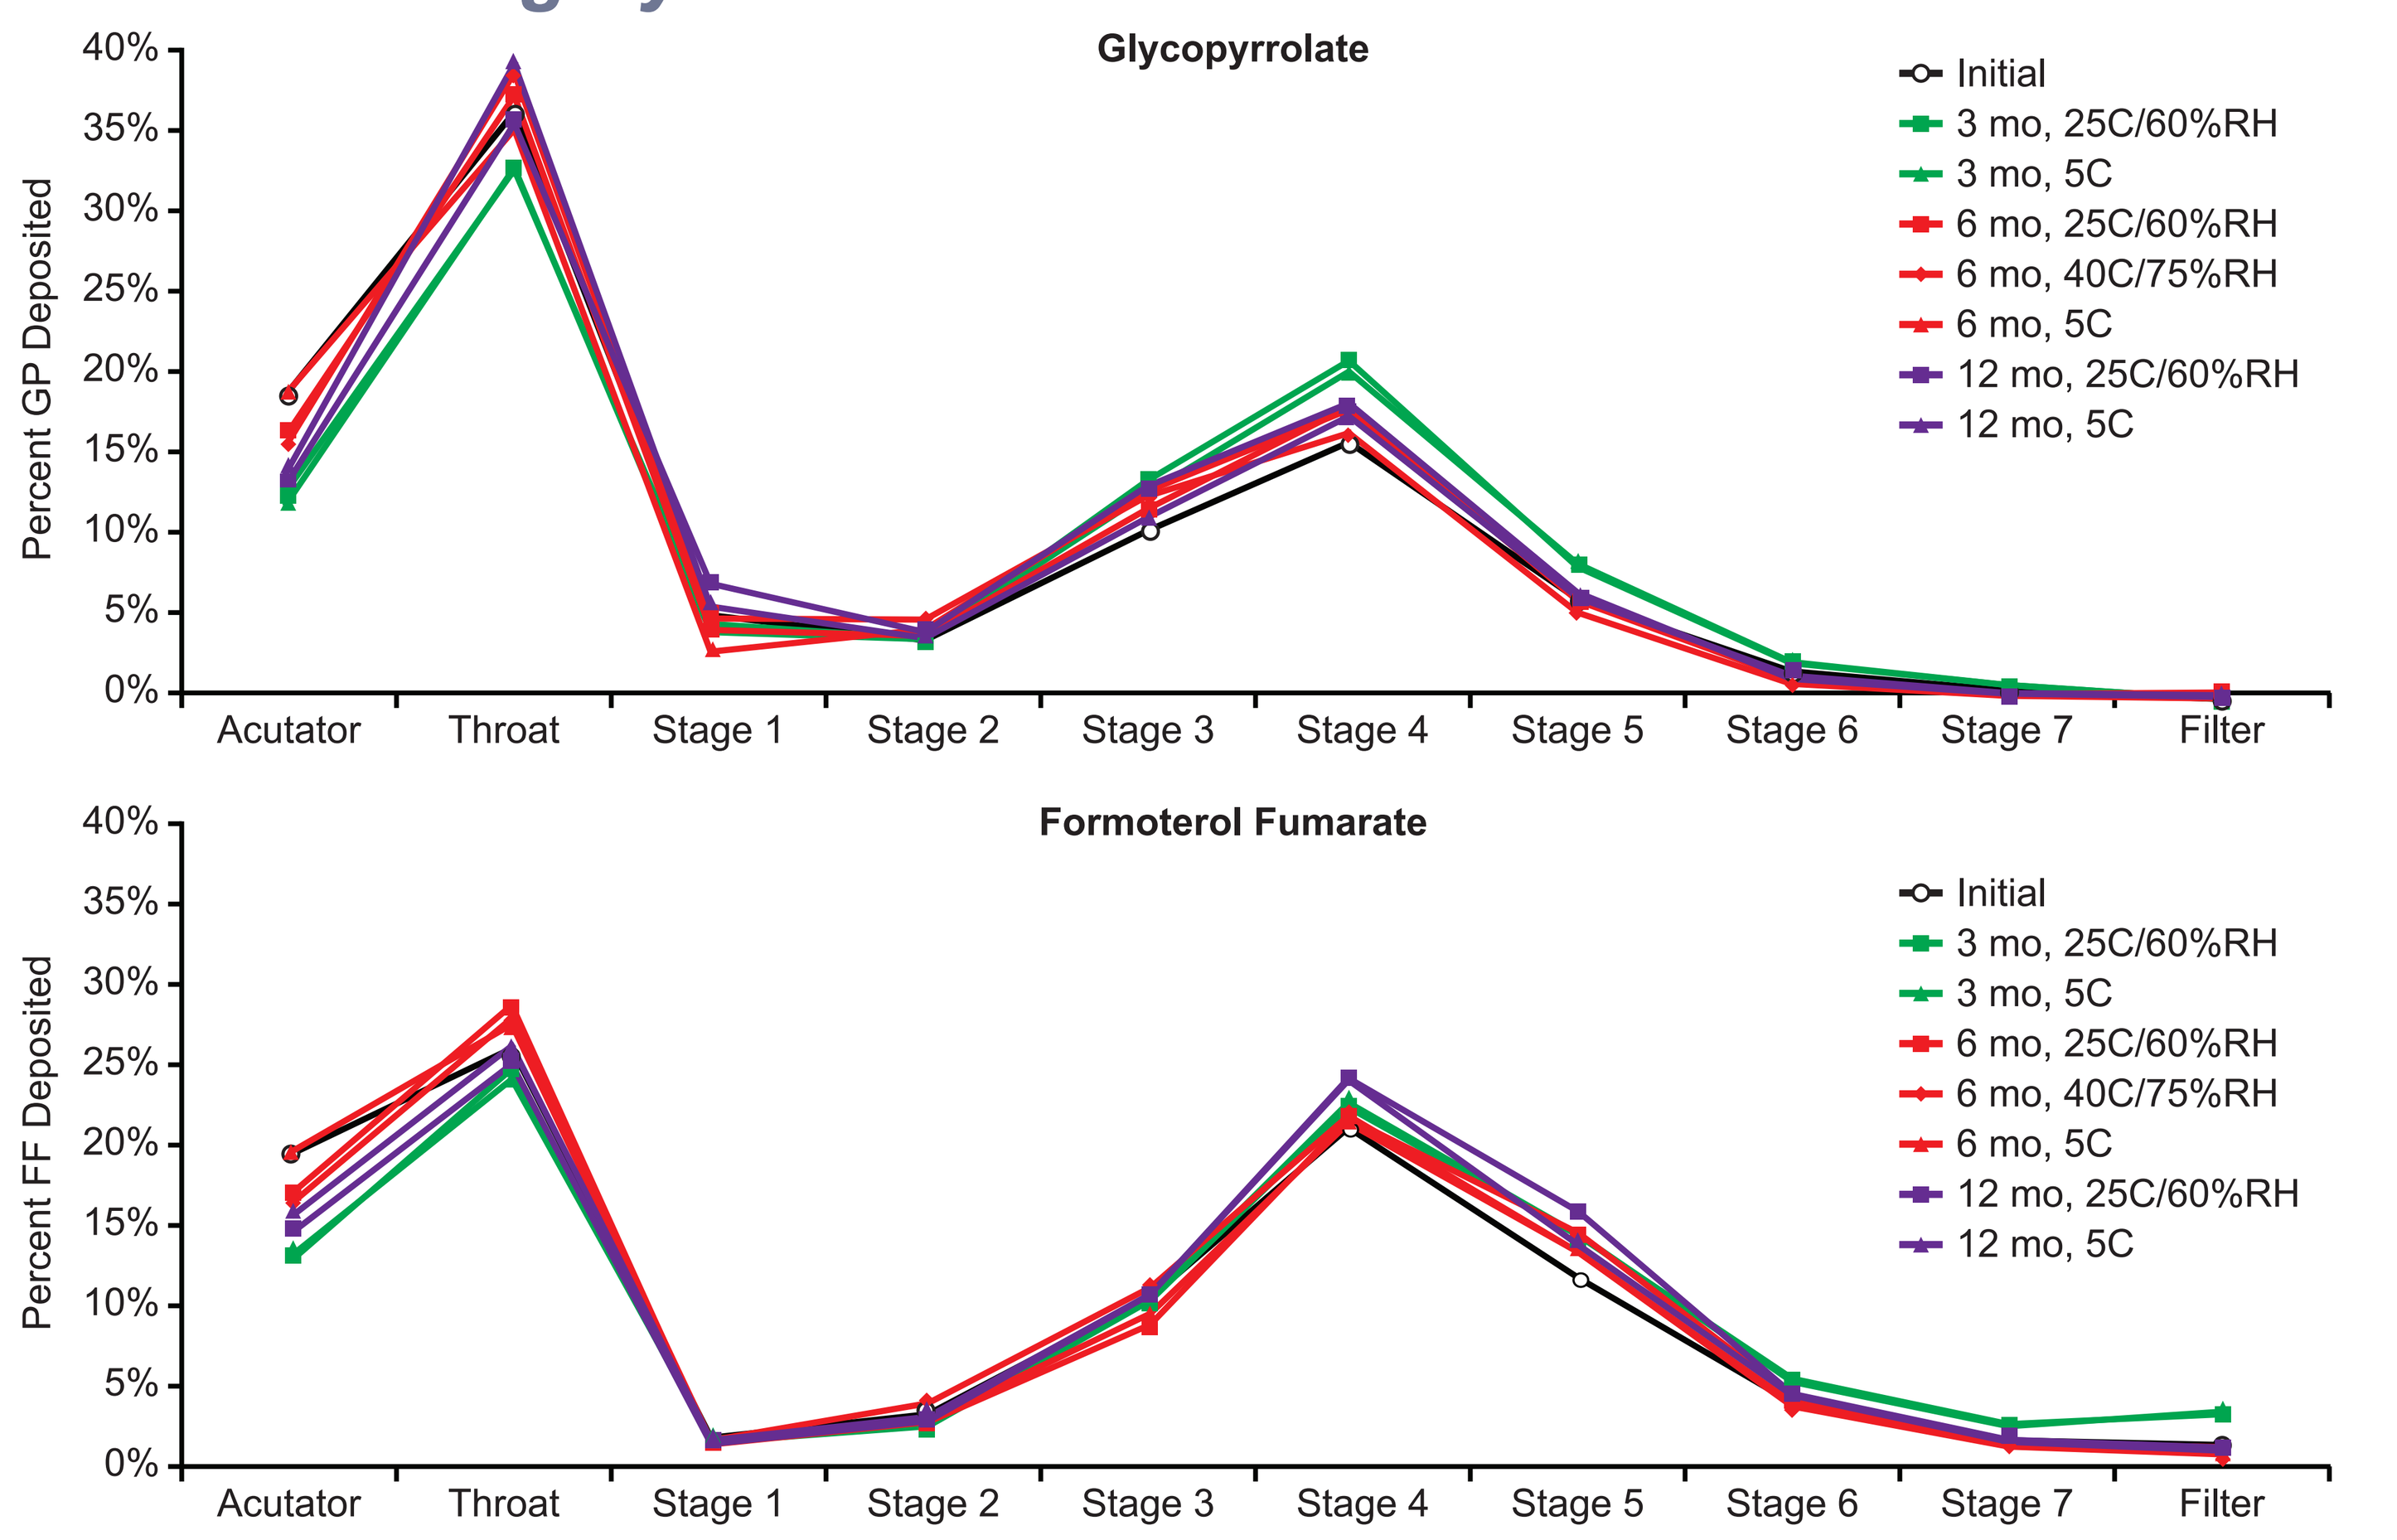
Glycopyrrolate
Formoterol Fumarate
Percent GP Deposited
Percent FF Deposited
Acutator
Throat
Stage 1
Stage 2
Stage 3
Stage 4
Stage 5
Stage 6
Stage 7
Filter
Initial
3 mo, 25C/60%RH
3 mo, 5C
6 mo, 25C/60%RH
6 mo, 40C/75%RH
6 mo, 5C
12 mo, 25C/60%RH
12 mo, 5C

Figure 3. GP/FF MDI (18/2.4 µg) Particle Size Stability

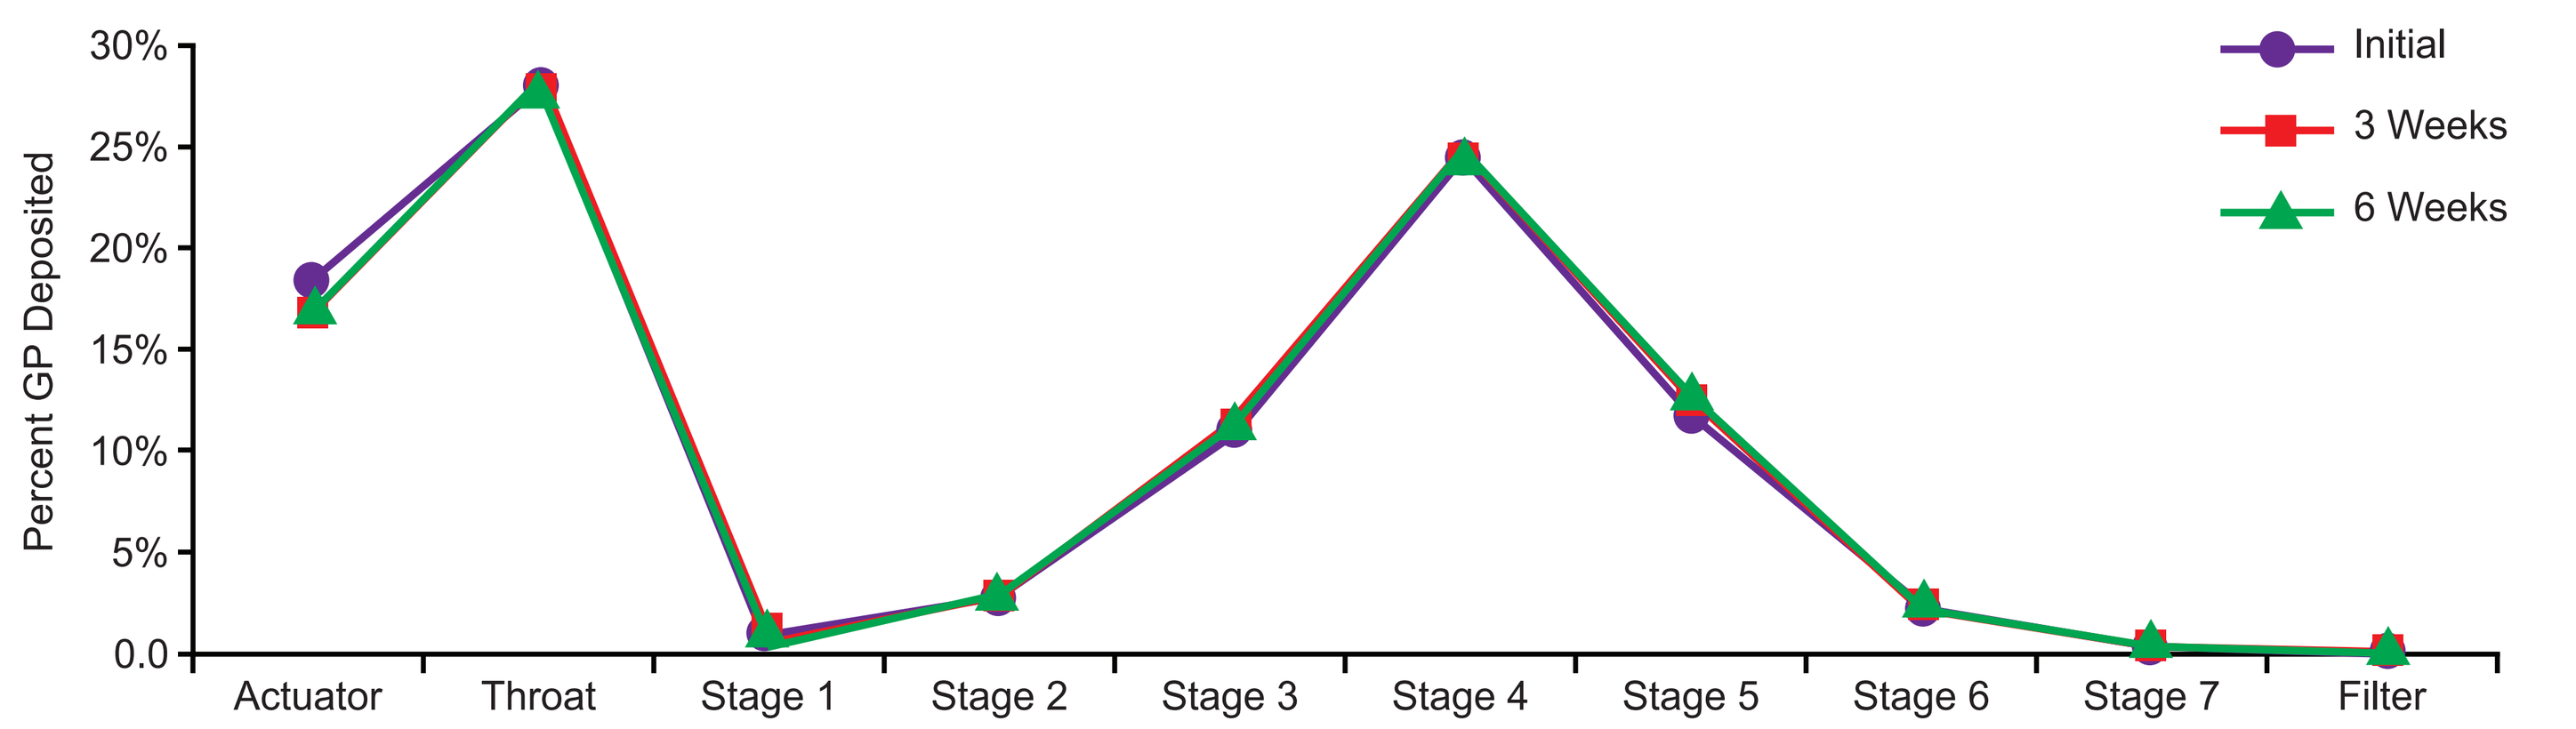
Percent GP Deposited
Actuator
Throat
Stage 1
Stage 2
Stage 3
Stage 4
Stage 5
Stage 6
Stage 7
Filter
Initial
3 Weeks
6 Weeks

Figure 4. GP MDI (4.5 µg) Temperature Cycling (-5 to 40ºC) Over Time

No Pharmaceutical Effect from Combination of APIs

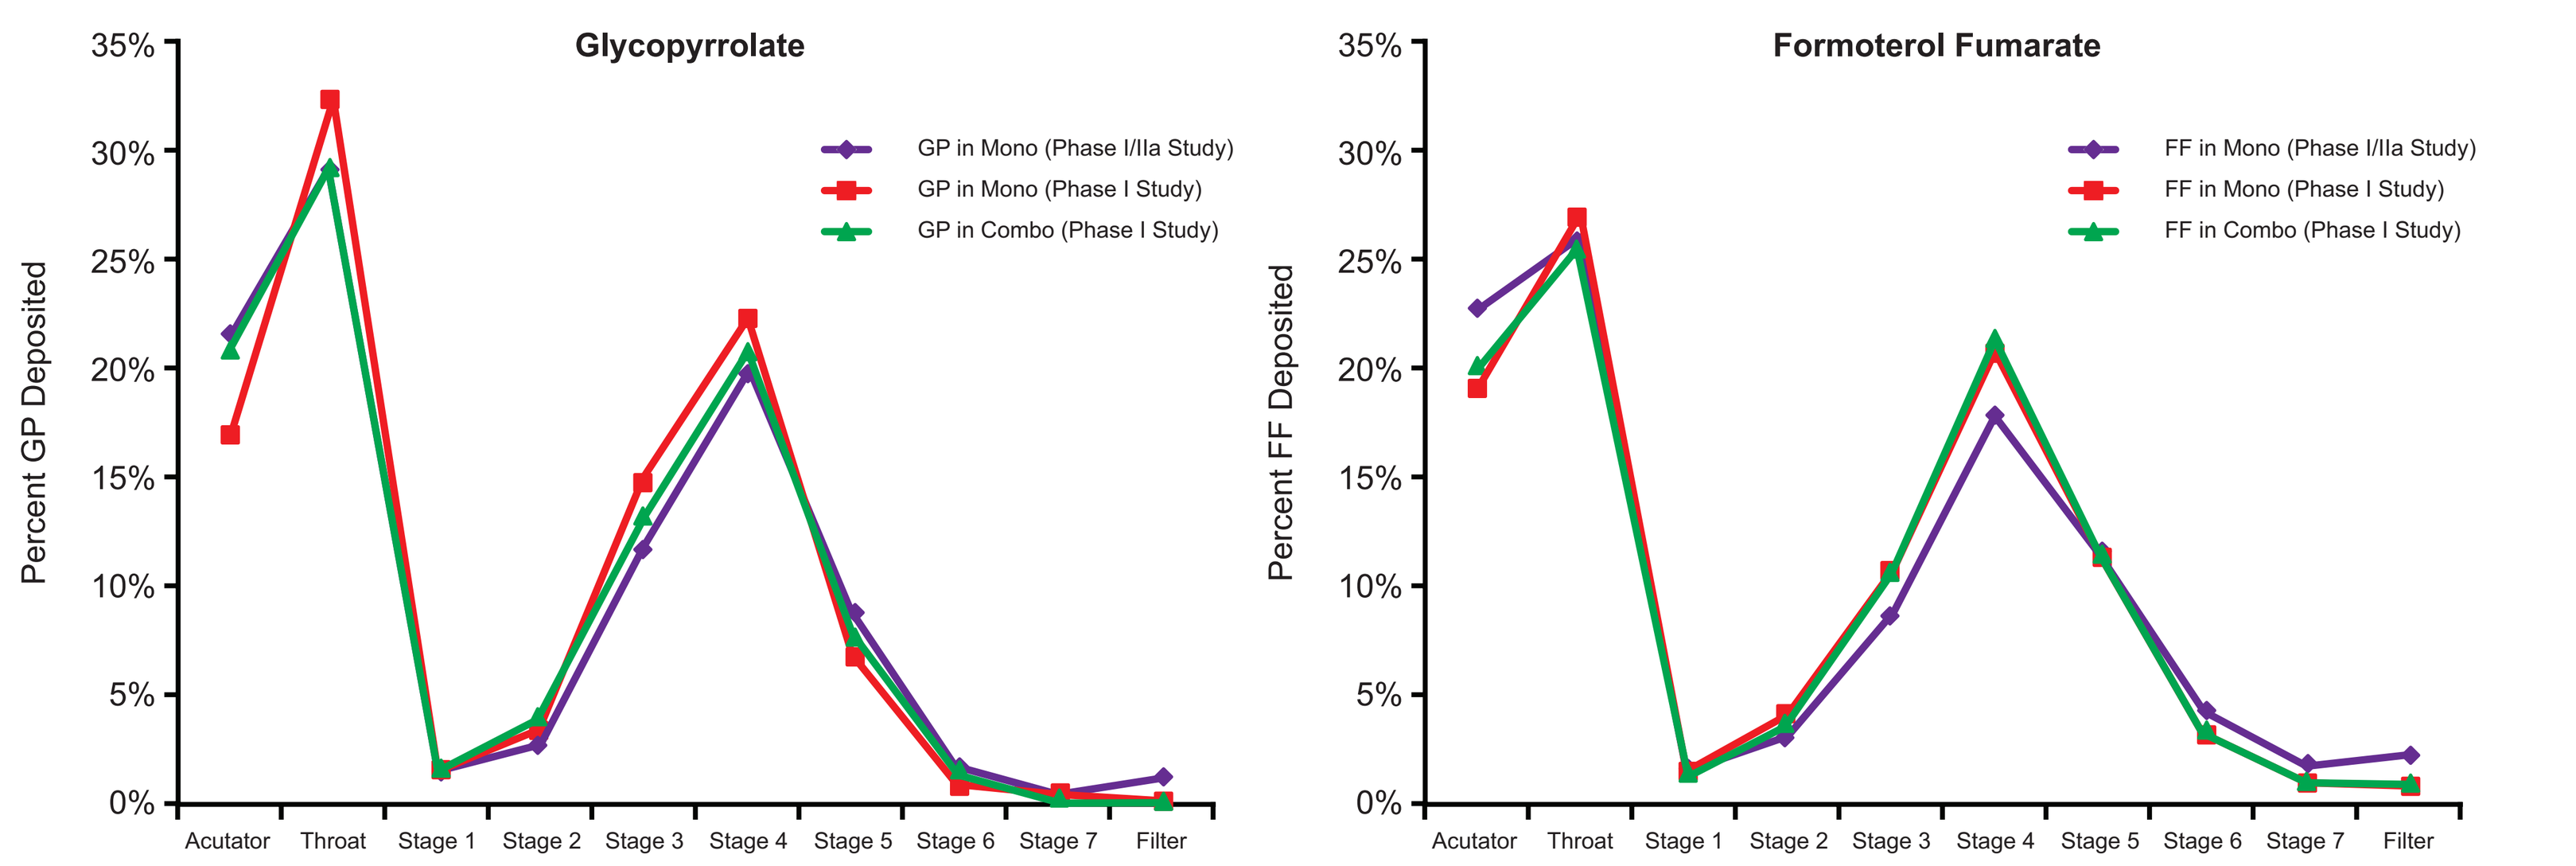
Glycopyrrolate
Formoterol Fumarate
Percent GP Deposited
Percent FF Deposited
GP in Mono (Phase I/IIa Study)
GP in Mono (Phase I Study)
GP in Combo (Phase I Study)
FF in Mono (Phase I/IIa Study)
FF in Mono (Phase I Study)
FF in Combo (Phase I Study)

Figure 5. GP MDI (18 µg), FF MDI (2.4 µg), and GP/FF MDI (18/2.4 µg)

API (Product)	%FPF	FPM (µg)	MMAD (µm)	% USP Throat
GP in mono (GP MDI, 18 µg)	55	11.4	3.5	32
GP in combo (GP/FF MDI, 18/2.4 µg)	56	10.6	3.4	30
FF in mono (FF MDI, 2.4 µg)	59	1.6	3.0	27
FF in combo (GP/FF MDI, 18/2.4 µg)	62	1.6	3.0	26

FPF = fine particle fraction; FPM = fine particle mass; MMAD = mass median aerodynamic diameter

Table 1. GP MDI (18 µg), FF MDI (2.4 µg), and GP/FF MDI (18/2.4 µg) Cascade Impaction Data

Chemically Stable

- No degradation of glycopyrrolate seen at any condition. No significant degradation of formoterol fumarate seen at room temperature.

Conclusions

Pearl is well positioned to further develop its MDI products. Pearl's MDI products demonstrate the following characteristics:

- Physical and chemical stability even under stressed conditions across a wide range of products
- Ability to develop very low doses of potent molecules
- High fine particle fraction (>50%) with low throat deposition for all products
- Excellent dose content uniformity
- No pharmaceutical effect observed when developing combination drug products
- High speed of development; < 9 months from first formulation to dosing patients

Pearl's porous particle platform is ideal for the development of robust MDI products in timelines not previously attainable.

References

1. Image of valves/actuators from Bespak website; image of cans from Presspart website.
2. Dellamary LA, Tarara TE, Smith DJ, Woelk CH, Adractas A, Costello ML, Gill H, Weers JG. Hollow porous particles in metered dose inhalers. *Pharm Res.* 2000 Feb;17(2):168-74.