Novel Experimental Method Indicates Proteins and Peptides Are Protected from High Gas Temperatures During Spray Drying

Reinhard Vehring, Vathana Tep, and Willard R. Foss
Nektar Therapeutics, San Carlos, CA

NEKTAR™

ABSTRACT

We studied the temperature history of droplets under typical spray-drying conditions for the production of respirable pharmaceutical powders.

A novel experimental method allows the direct measurement of droplet evaporation rates. The experimental results are compared to a numerical model of the spray-drying process to determine the droplet temperature.

The results show that during spray drying of respirable particles, the active pharmaceutical ingredients are protected by evaporative cooling. The particles remain near room temperature until the evaporation is nearly complete, at which time the active ingredients are at a sufficiently high concentration to be immobilized and protected in a glassy matrix.

OBJECTIVES

- To assess the temperature stress on active pharmaceutical ingredients during spray drying
- To provide experimental and numerical analysis of the drying process

INTRODUCTION

Experiments on single suspended droplets have indicated that high temperatures in the liquid phase can occur during droplet drying in a hot gas stream[1]. It has been suggested that droplets heat up to similarly high temperatures during spray-drying processes with a potentially detrimental effect on heat-sensitive active pharmaceutical ingredients.

Studies on the evaporation behavior of solution droplets have so far been restricted to conditions that are atypical of the spray-drying of powders intended for pulmonary delivery. The resulting dry particles were orders of magnitude larger than respirable particles.

The methods presented here allow measurement of the evaporation process under realistic conditions. The apparatus produces model particles with a similar morphology and size to those found in the actual dry powder product.

METHODOLGY

Experimental

Monodisperse droplets are produced by a droplet-on-demand generator and injected into a laminar gas flow of controlled temperature, velocity, and relative humidity. Initial droplet diameters are between 10 and 30 µm.

The droplets move along a highly repeatable trajectory and can be sized at different distances, z, from the point of injection. The distance a droplet travels as a function of time is measured using stroboscopic illumination. Thus, the droplet diameter as a function of time can be determined for evaporating droplets.

2003 AAPS Annual Meeting and Exposition, October 26-30, 2003,
Salt Lake City, Utah

The size of the droplets, the time-scale of the process, and the gas phase conditions are representative of typical spray-drying conditions. Droplets are spaced far enough apart to avoid droplet-droplet interactions.

Figure 1. Production of Representative Model Droplets in a Controlled Gas Phase

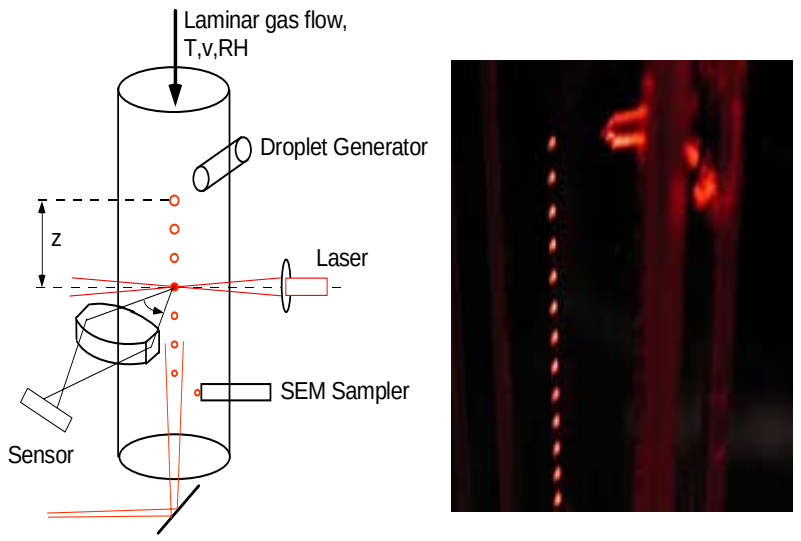
Laminar gas flow,
T,v,RH
Droplet Generator
z
Laser
SEM Sampler
Sensor

The droplet diameter is determined by measuring the average peak separation in the phase function of the elastical light scattering. The correlation between peak separation and droplet size was established with a numerical light-scattering model using Mie theory.

Figure 2. Droplet Sizing Using a Laser Light-Scattering Method

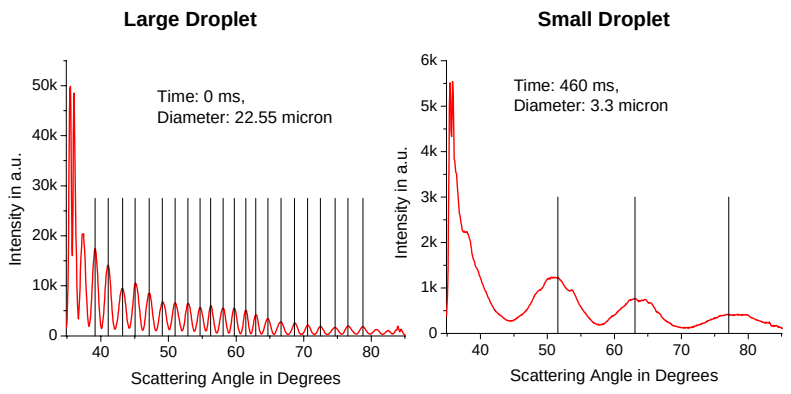
Large Droplet
Time: 0 ms,
Diameter: 22.55 micron
Intensity in a.u.
Scattering Angle in Degrees
Small Droplet
Time: 460 ms,
Diameter: 3.3 micron
Intensity in a.u.
Scattering Angle in Degrees

Numerical Model

A numerical model was developed that describes the radial distribution of all substances and the temperature in a multicomponent droplet. Both the liquid and the gas phases are modeled. The model is spherically symmetric and transient. It allows for variable material properties and a moving gas-liquid boundary. The governing equations are given below:

Weight fraction of species i (w_i) in droplet and gas:

$$\rho\frac{\partial w_i}{\partial t}+\rho v\frac{\partial w_i}{\partial r}=\frac{1}{r^2}\frac{\partial}{\partial r}\left(r^2\rho D_i\frac{\partial w_i}{\partial r}\right)\quad\text{and}\quad\sum_{i=1}^{n}w_i=1$$

Weight fractions at gas/droplet interface:

$$\left[\rho D_i\frac{\partial w_i}{\partial r}+\rho w_i\left(\frac{\partial R}{\partial t}-v\right)\right]_R^{(\text{drop})}=\left[\rho D_i\frac{\partial w_i}{\partial r}+\rho w_i\left(\frac{\partial R}{\partial t}-v\right)\right]_R^{(\text{gas})}$$

and $a_i(w_i)\big|_R^{(\text{drop})}=\dfrac{p_i}{p_i^{sat}}\Big|_R^{(\text{gas})}$ where a_i is the activity of species i

Droplet size (R) and radial velocity (v) in droplet and gas:

$$\frac{\partial R}{\partial t}=\left[\frac{(\rho v)^{(\text{drop})}-(\rho v)^{(\text{gas})}}{\rho^{(\text{drop})}-\rho^{(\text{gas})}}\right]_R\quad\text{and}\quad v=-\frac{1}{r^2\rho}\int_0^r\xi^2\frac{\partial\rho}{\partial t}d\xi$$

Gas temperature (T):

$$\rho\hat{C}_p\frac{\partial T}{\partial t}+\rho\hat{C}_p v\frac{\partial T}{\partial r}=\frac{1}{r^2}\frac{\partial}{\partial r}\left(r^2k\frac{\partial T}{\partial r}\right)+\sum_{i=1}^{n}\left(\overline{H}_i\frac{1}{r^2}\frac{\partial}{\partial r}\left(r^2\rho D_i\frac{\partial w_i}{\partial r}\right)\right)$$

Droplet temperature (T_{drop}):

$$\frac{R\rho C_p}{3}\frac{\partial T_{\text{drop}}}{\partial t}=\left[k\frac{\partial T}{\partial r}+\rho\sum_{i=1}^{n}\overline{H}_iD_i\frac{\partial w_i}{\partial r}+\rho\overline{H}\left(\frac{\partial R}{\partial t}-v\right)\right]_R^{(\text{gas})}+\rho^{(\text{drop})}\Delta H_{vap}\frac{\partial R}{\partial t}$$

Method Verification

The validity of the model and the experimental method was verified with experiments on pure water droplets.

Figure 3. Experimental Determination of Evaporation Rates

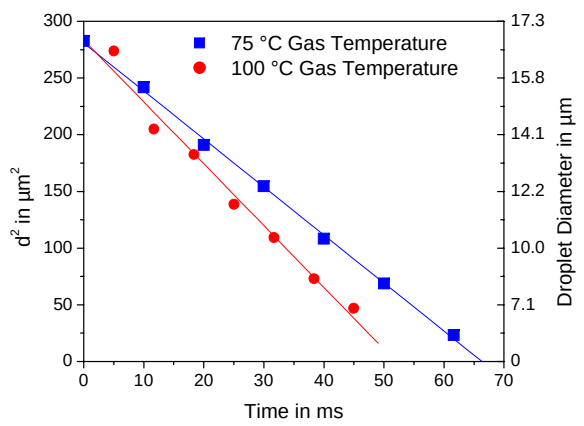
75 °C Gas Temperature
100 °C Gas Temperature
d² in µm²
Droplet Diameter in µm
Time in ms

Figure 4. Good Agreement Between Experimental Results and Numerical Model

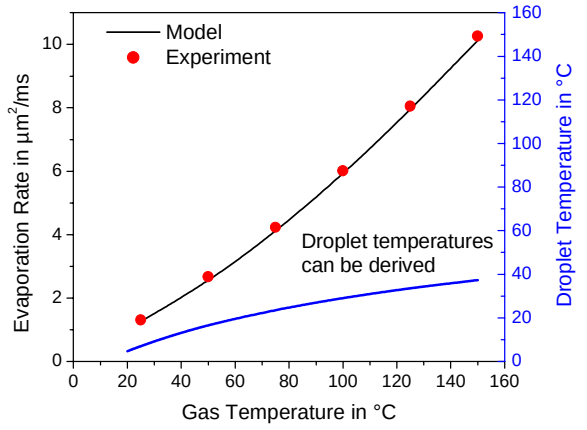
Model
Experiment
Droplet temperatures
can be derived
Evaporation Rate in µm²/ms
Droplet Temperature in °C
Gas Temperature in °C

Figure 5. Model Particles with Representative Morphologies

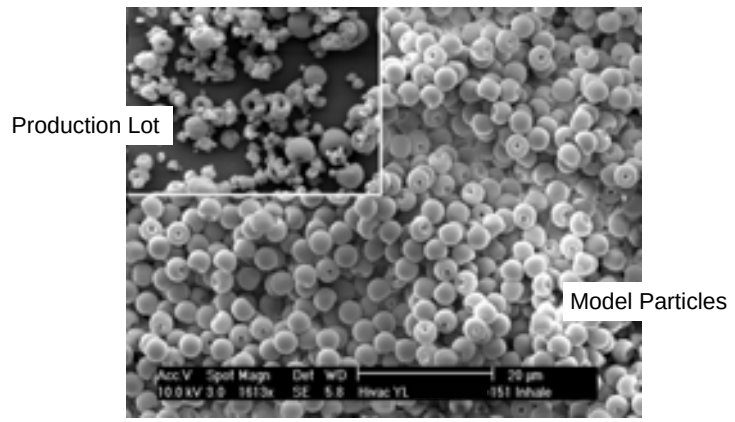
Production Lot
Model Particles

The experimental technique adequately models the real process.

RESULTS

Figure 6. Spray Drying of a Glycoprotein

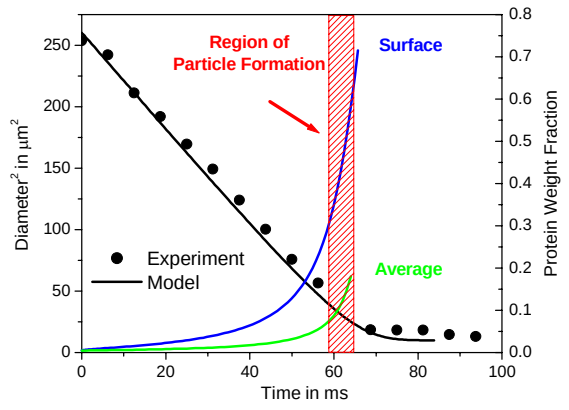
Region of
Particle Formation
Surface
Average
Experiment
Model
Diameter² in µm²
Protein Weight Fraction
Time in ms

Figure 7. Temperature Stress on the Protein During Manufacturing and Storage

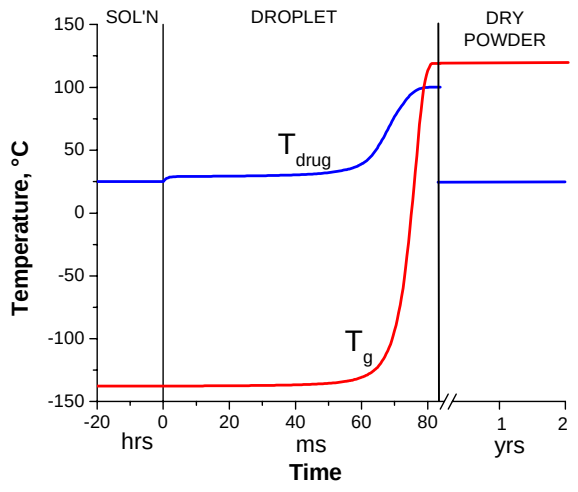
SOL'N
DROPLET
DRY
POWDER
T drug
T g
Temperature, °C
hrs
ms
yrs
Time

CONCLUSIONS

- Respirable particles are typically formed in less than 100 ms.
- During the evaporation, the active pharmaceutical ingredients are protected from high temperature by evaporative cooling.
- Shell formation on micron-sized droplets does not lead to a significant decrease in evaporation rate. The droplets do not boil or explode.
- When the particles are almost dry, their temperature will reach the drying gas temperature for fractions of a second but the proteins or peptides are by then immobilized and protected in an amorphous phase.

Spray-drying subjects proteins or peptides to only minor temperature stress, which generally does not cause significant degradation. It is a well suited manufacturing process for temperature sensitive active pharmaceutical ingredients.

REFERENCES

1. J.-C Lin and J. W. Gentry, Aerosol Science and Technology, 2003, 37:15-32.

ACKNOWLEDGMENTS

The authors are grateful for the support and assistance of John WY Lee, Nazli Egilmez, David Lechuga, and Dan Miller.