Peptide Stability in Spray-Dried Powders for Inhalation

Hak-Kim Chan[2], David Lechuga-Ballesteros[1], S. Russ Lehrman[1], Danforth P. Miller[1]
Katherine Pikal-Cleland[1], Trixie Tan[1], Reinhard Vehring[1], and Lisa A. Williams[1]

[1]Inhale Therapeutic Systems, Inc., San Carlos, California [2]Faculty of Pharmacy, University of Sydney University, Sydney, Australia

OBJECTIVE

To identify experimental parameters that affect the solid-state aggregation of salmon calcitonin (sCT).

INTRODUCTION

Salmon calcitonin (sCT) is a basic, 32-residue peptide that increases Ca^{++} uptake and is useful in the treatment of osteoperosis. As with the human homolog, this peptide is known for its ability to aggregate when solubilized at high concentration (>1 mg/mL), room temperature, physiological pH and ionic strength.[1,2]

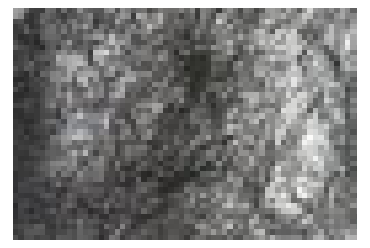

NMR data suggests that in solution, monomeric calcitonin forms a metastable α-helix.[3] Fibrillation occurs upon association of these monomers and the adoption of a β-sheet conformation.[3] We are interested in understanding if sCT aggregates form in the solid state and, if so, experimental parameters that affect this process.

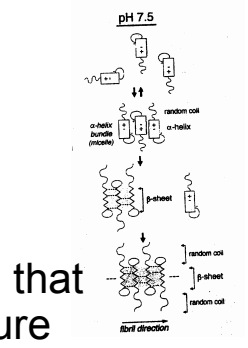

pH 7.5
fibril direction

Chemical stability in dry powders is often affected by factors that affect molecular mobility.[4,5] For example, lowering temperature and relative humidity (RH) usually discourage degradative processes. Excipients that have high T_g also help stabilize proteins in the solid-state by reducing mobility.[6] This study, therefore, investigates the physical stability of sCT as a function of temperature, RH, peptide and sugar concentration.

MATERIALS & METHODS

• Salmon calcitonin used in these studies was obtained from Bachem AG (Torrance, CA). Stability samples were stored at 25°C at the relative humidities in the text.

• Aqueous solutions containing sCT:mannitol ratios of 0:100, 10:90, 30:70, 50:50, 70:30, 90:10, and 100:0 (pH 7) were spray-dried using a Büchi 190 spray dryer. Inlet and outlet spray dryers were 125° and 70°C.

• Aggregation of processed and stored samples was monitored by SEC. A Tosohaas TSK-Gel G2000SW_{XL} column was eluted with 0.25 M Na_2SO_4 at a flow rate of 0.7 mL/min (25°C). Column effluent was monitored at 210nm.

• Chemical stability was monitored by RP-HPLC. A Vydac C18 column was eluted with a CH_3CN gradient (0.1% TFA) at a flow rate of 1 mL/min (40°C). Column effluent was monitored at 210nm.

• Raman spectra were obtained in order to determine the extent of powder crystallinity. Laser light (669.85nm, 100 mW) used to irradiate the powder, which was maintained at 22°C, 0% RH. Raman scatter was collected at a 90° angle. Spectral resolution was 3 cm^{-1}; positional accuracy was1 cm^{-1}.

• Powder hygroscopicity was assessed by using a dynamic vapor sorption (DVS) microbalance (Surface Measurement Systems, UK). Samples were thoroughly dried at the beginning of each experiment. RH was varied from 0 to 90% RH in increments of 5% RH, and from 90 to 0% RH in increments of 10% RH.

• Interactions of powder with water were measured using isothermal microcalorimetry with an RH perfusion cell (2277 Thermal Activity Monitor, TAM, Thermometric AB, Sweden).

• FTIR was performed on a ThermoNicolet Magna 760 IR. Samples (0.2-0.4 mg) were prepared as KBr pellets (300 mg). Spectra were obtained at 4 cm^{-1} resolution (25°C).

RESULTS AND DISCUSSION

1 HPLC analysis of spray-dried powders shows that chemical modification and peptide aggregation is not significant.

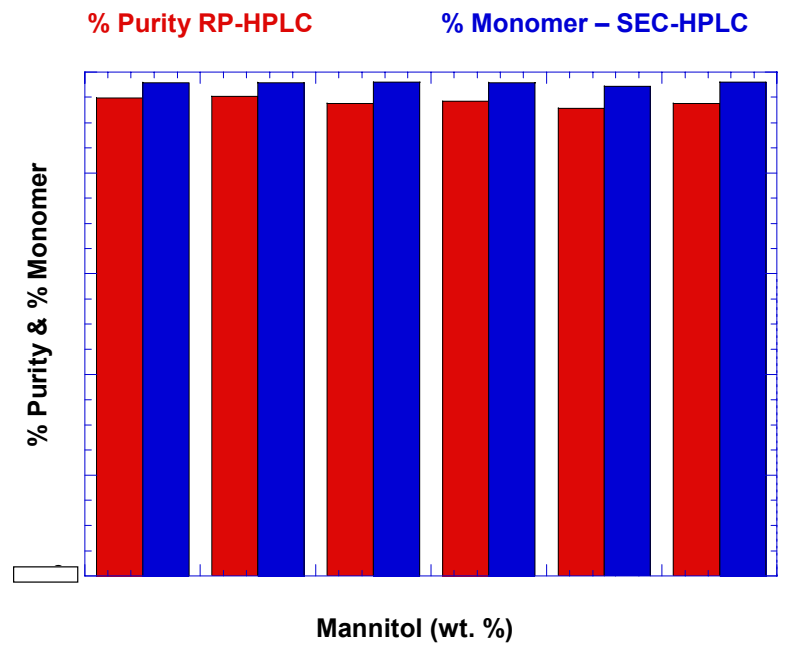

% Purity RP-HPLC
% Monomer – SEC-HPLC
% Purity & % Monomer
Mannitol (wt. %)

2 Following storage at 29 and 84% RH for 5 days, powders containing 70% sCT contain a soluble aggregate.

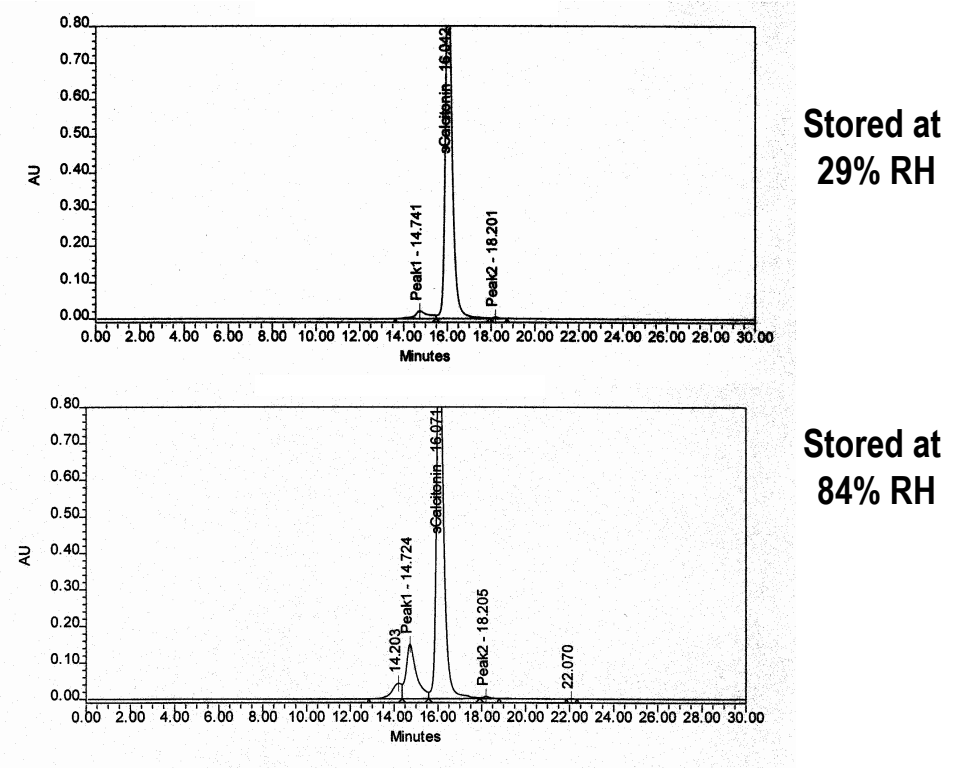

Stored at 29% RH
Stored at 84% RH
AU
Minutes
Peak1 - 14.741
sCalcitonin - 16.042
Peak2 - 18.201
14.203
Peak1 - 14.724
sCalcitonin - 16.071
Peak2 - 18.205
22.070

3 sCT aggregation increases when sCT content or relative humidity is increased.

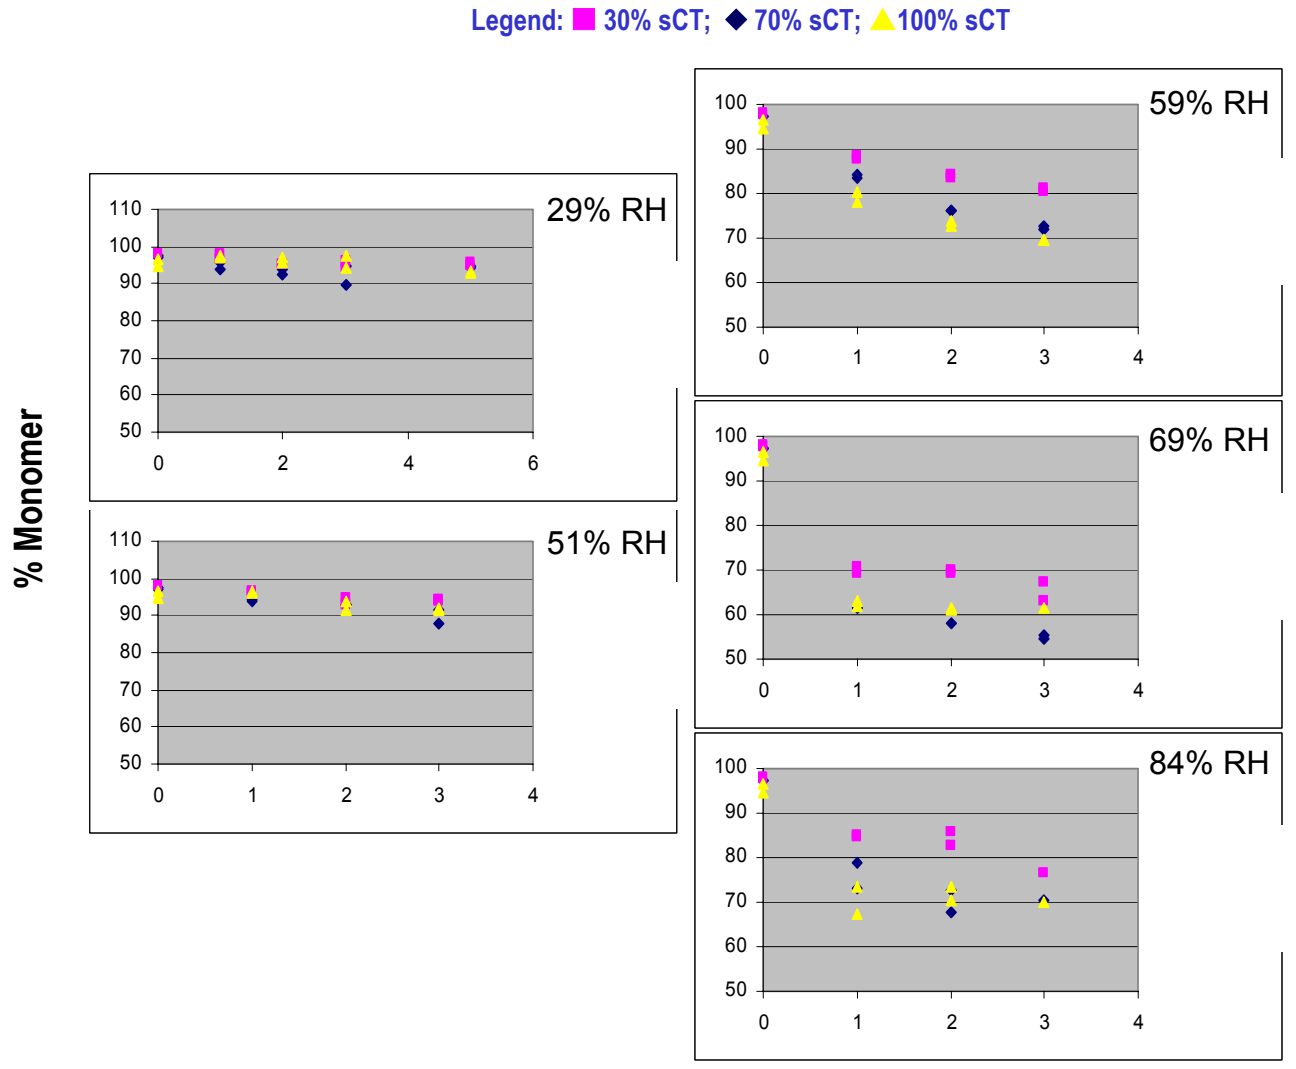

Legend: 30% sCT; 70% sCT; 100% sCT
% Monomer
29% RH
51% RH
59% RH
69% RH
84% RH

4 Increasing the sCT content of these powders increases their capacity for moisture.

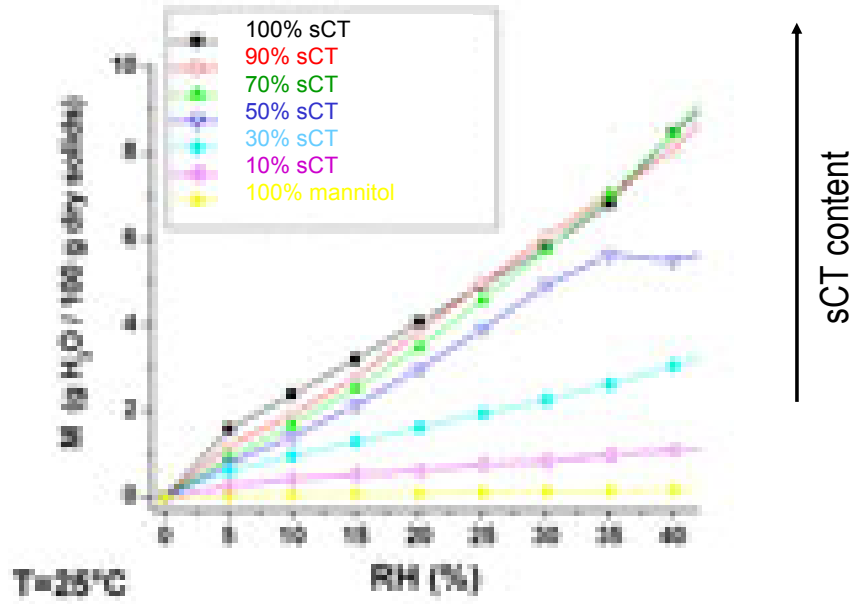

100% sCT
90% sCT
70% sCT
50% sCT
30% sCT
10% sCT
100% mannitol
sCT content
T=25°C
RH (%)

5 FTIR studies show that storing high sCT content powders at high RH promotes the formation of b-sheet conformation.

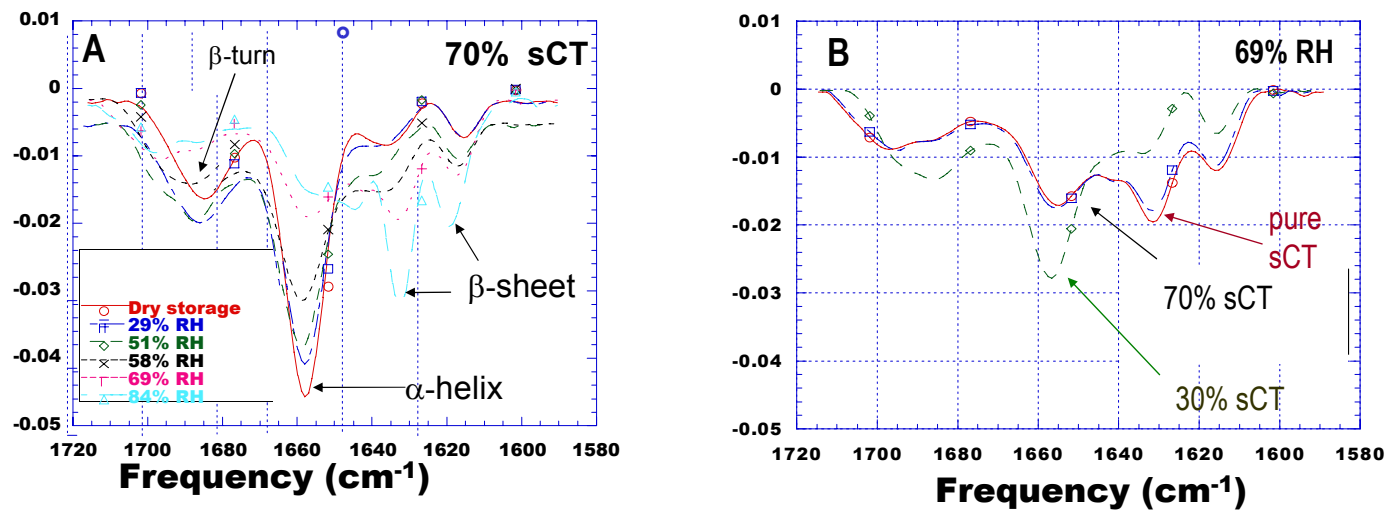

A
β-turn
70% sCT
β-sheet
α-helix
Dry storage
29% RH
51% RH
58% RH
69% RH
84% RH
Frequency (cm-1)
B
69% RH
pure sCT
70% sCT
30% sCT
Frequency (cm-1)

6 Dynamic water sorption studies, performed at 25° C, show that high sCT content powder absorbs more water than low sCT content powder.

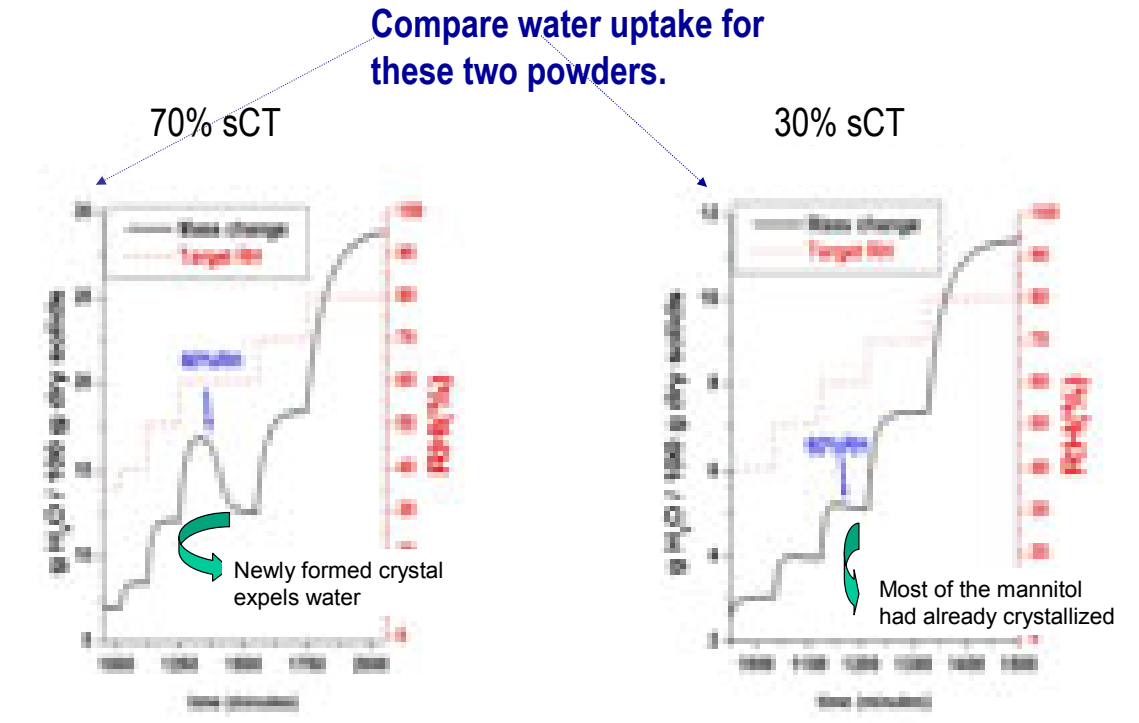

Compare water uptake for these two powders.
70% sCT
30% sCT
Newly formed crystal expels water
Most of the mannitol had already crystallized

Note the mannitol crystallization at 60% RH.

7 Raman spectra confirm that the powder containing 70% mannitol (i.e., 30% sCT) is crystalline from 0 to 84% RH. Powders containing 30% mannitol crystallize only at high RH.

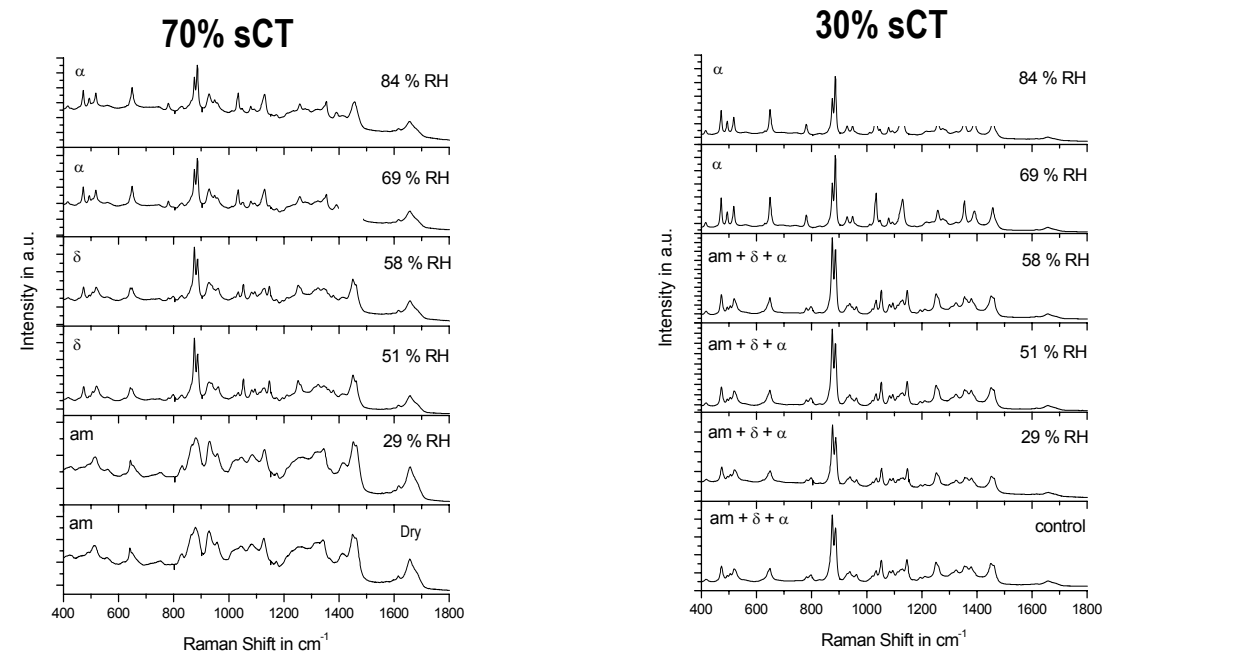

70% sCT
30% sCT
84 % RH
69 % RH
58 % RH
51 % RH
29 % RH
Dry
control
Intensity in a.u.
Raman Shift in cm-1

5 days, 25° C

8 Moisture induced thermal activity trace reveals the critical RH for mobility (RH_p) and crystallization RH (RH_x). Performing this experiment at 25 °, 40 ° and 60° C facilitates identification of danger and safe storage zones.

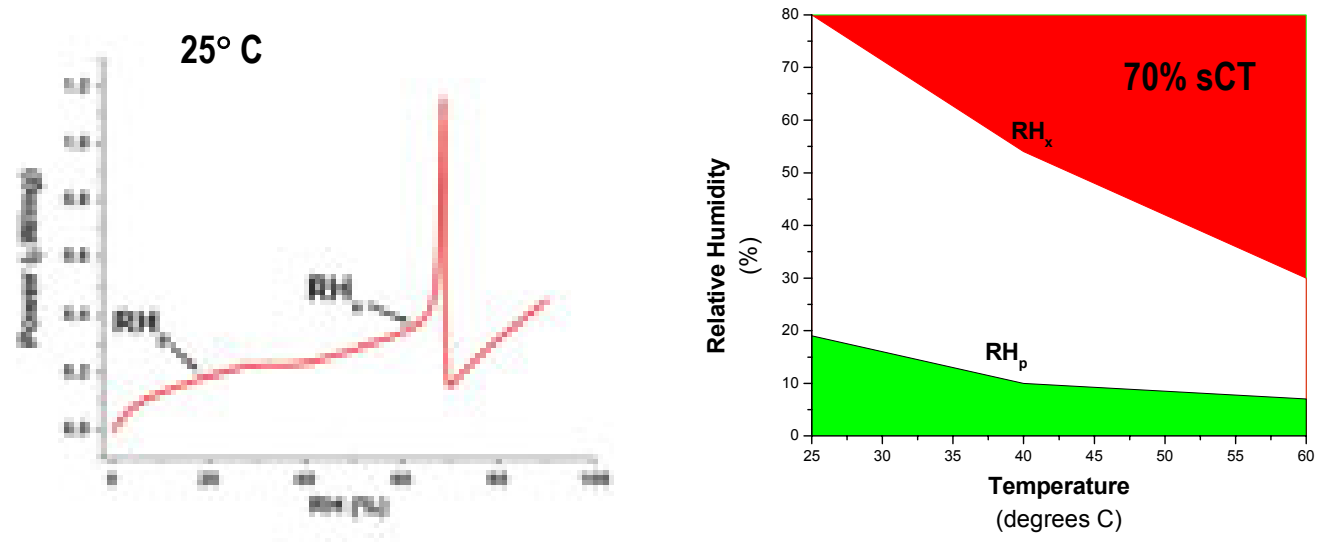

25° C
70% sCT
RHx
RHp
Relative Humidity (%)
Temperature (degrees C)

CONCLUSIONS

- No sCT degradation is observed when this peptide is spray-dried using the conditions described here. However, powders stored at high RH aggregate more rapidly than those stored at low humidity.
- High humidity parallels changes in sCT conformation, particularly those powders containing high sCT content. High RH and sCT content promote β-sheet formation. However, no clear relationship between powder crystallinity and aggregation was observed.
- In these spray-dried powders, sCT aggregation can be controlled by limiting moisture and peptide content.

ACKNOWLEDGMENTS

The authors are grateful for the contributions of Chatan Charan, Sarma Duddu, Mei-Chang Kuo, and Herm Snyder of Inhale Therapeutic Systems, Inc.

REFERENCES

1. Gilchrist PJ, Bradshaw JP, *Biochim Biophys Acta* (1993) 1182:111-114.
2. Arvinte T, Cudd A, Drake AF, *J. Biological Chemistry* (1992) 268: 6415-6422.
3. Kamhira M, *et al., Protein Science* (2000) 9:867-877.
4. Lai MC, and Topp EM, *J. of Pharmaceutical Sciences* (1999) 88:489-500.
5. Pikal MJ, *et. al.*, *Pharmaceutical Research* (1991) 8:*427-436.*
6. Duddu SP, Zhang G, Dal Monte PR, *Pharmaceutical Research* (1997) 14:596-600.

Presented at 2002 AAPS Annual Meeting and Exposition, Nov. 10 – 14, 2002, Toronto, Ontario, Canada